EXPLORING THE MULTIPLE MECHANISMS UNDERLYING HER-096 NEUROPROTECTION AND REGENERATION

Natalia Kulesskaya, Kira M. Holmström and Henri J. Huttunen

Herantis Pharma Plc, Espoo, Finland

SUMMARY

HER-096 is a brain-penetrating peptidomimetic derived from the active site of the unconventional neurotrophic factor CDNF, and is developed as a disease-modifying treatment for Parkinson's disease. HER-096 shows similar potency and mechanism of action as CDNF and promotes neuroregeneration restoring proteostasis and reducing neuroinflammation in animal model of synucleopathy [1].

The neuroprotective actions of CDNF and HER-096 are mainly mediated via modulation of the endoplasmic reticulum (ER) stress response and unfolded protein response (UPR) pathway. However, there are also effects on glial and immune cells, and the relative contribution of different cell types to the overall therapeutic effect is incompletely understood. Here, we further explored the effects of HER-096 in an aged mouse model of synucleinopathy and with human iPSC motor neurons.

[1] Kulesskaya N, Bhattacharjee A, Holmström KM, Vuorio P, Henriques A, Callizot N, Huttunen HJ. HER-096 is a CDNF-derived brain-penetrating peptidomimetic that protects dopaminergic neurons in a mouse synucleinopathy model of Parkinson's disease. Cell Chem Biol. 2023 Nov 24: S2451-9456(23)00420-8.

METHODS

Aged mouse model of synucleopathy is based on preformed alpha-synuclein fibril injection (bilateral substantia nigra) in combination with chronic glucocerebrosidase (GCase) inhibition by conduritol B-epoxide (CBE) in 18-month-old C57BL6 mice (Figure 1). HER-096 (10 mg/kg) was administered subcutaneously (s.c.) three times of week for 5 weeks starting from day 7 after model induction. Dopaminergic cell survival and effect on α-synuclein aggregation and phosphorylation was assessed by immunohistochemistry method (ICH). Neurotransmitter and metabolite from striatum and protein levels from lysate of substantia nigra were analysed by mass spectrometry and immunoassays. Transcriptomic changes were analysed from total RNA extracted from dissected striatum and substantia nigra tissue by Affymetrix Clariom D Mouse microarray. Gene Set Enrichment Analysis (GSEA) was used to assess changes in gene expression between treatment groups and timepoints.

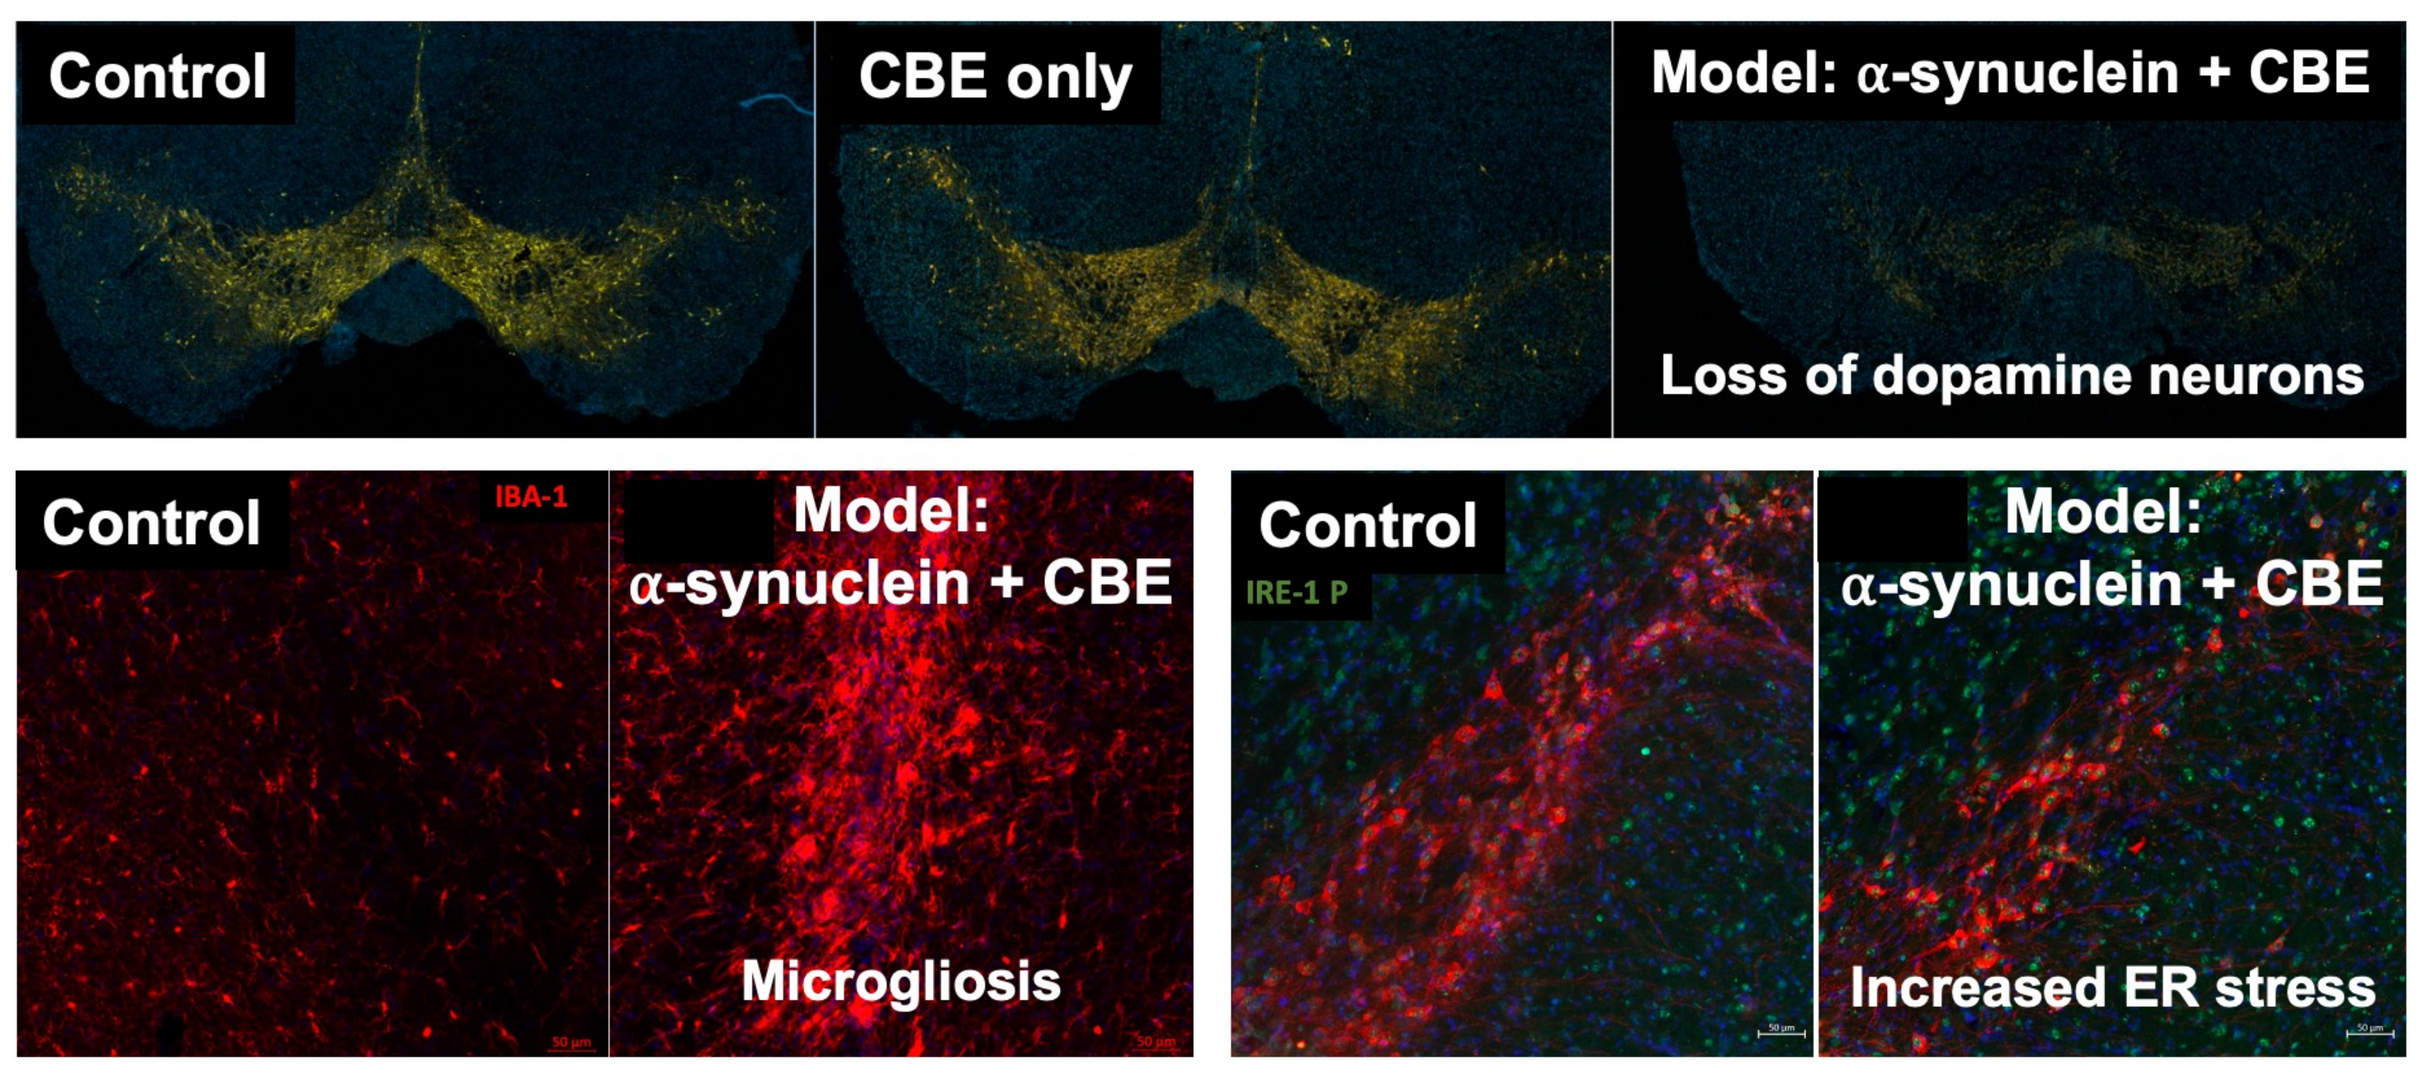
Control
CBE only
Model: α-synuclein + CBE
Loss of dopamine neurons
Control
IBA-1
Model:
α-synuclein + CBE
Microgliosis
Control
IRE-1 P
Model:
α-synuclein + CBE
Increased ER stress

Figure 1. *Representative images of immunohistochemical staining demonstrate the loss of dopamine neurons (tyrosine hydroxylase), increased microgliosis (Iba1) and ER stress (phospho-Ser724 IRE1) in mouse substantia nigra 4 weeks after intranigral α-synuclein protofibril injections and chronic GCase inhibition.*

In vitro regeneration of axotomized hiPSC motor neurons was done in microfluidic devices (ANANDA Devices). On day 10 after seeding, the motor neurons were axotomized by shear force created by vacuum aspiration. HER-096 was applied at 5 nM for 6 days starting from 4 hours before the axotomy. Seven-factor neuromorphological analysis was performed to assess the neuroregenerative effects of HER-096 on axotomized motor neurons (Figure 2).

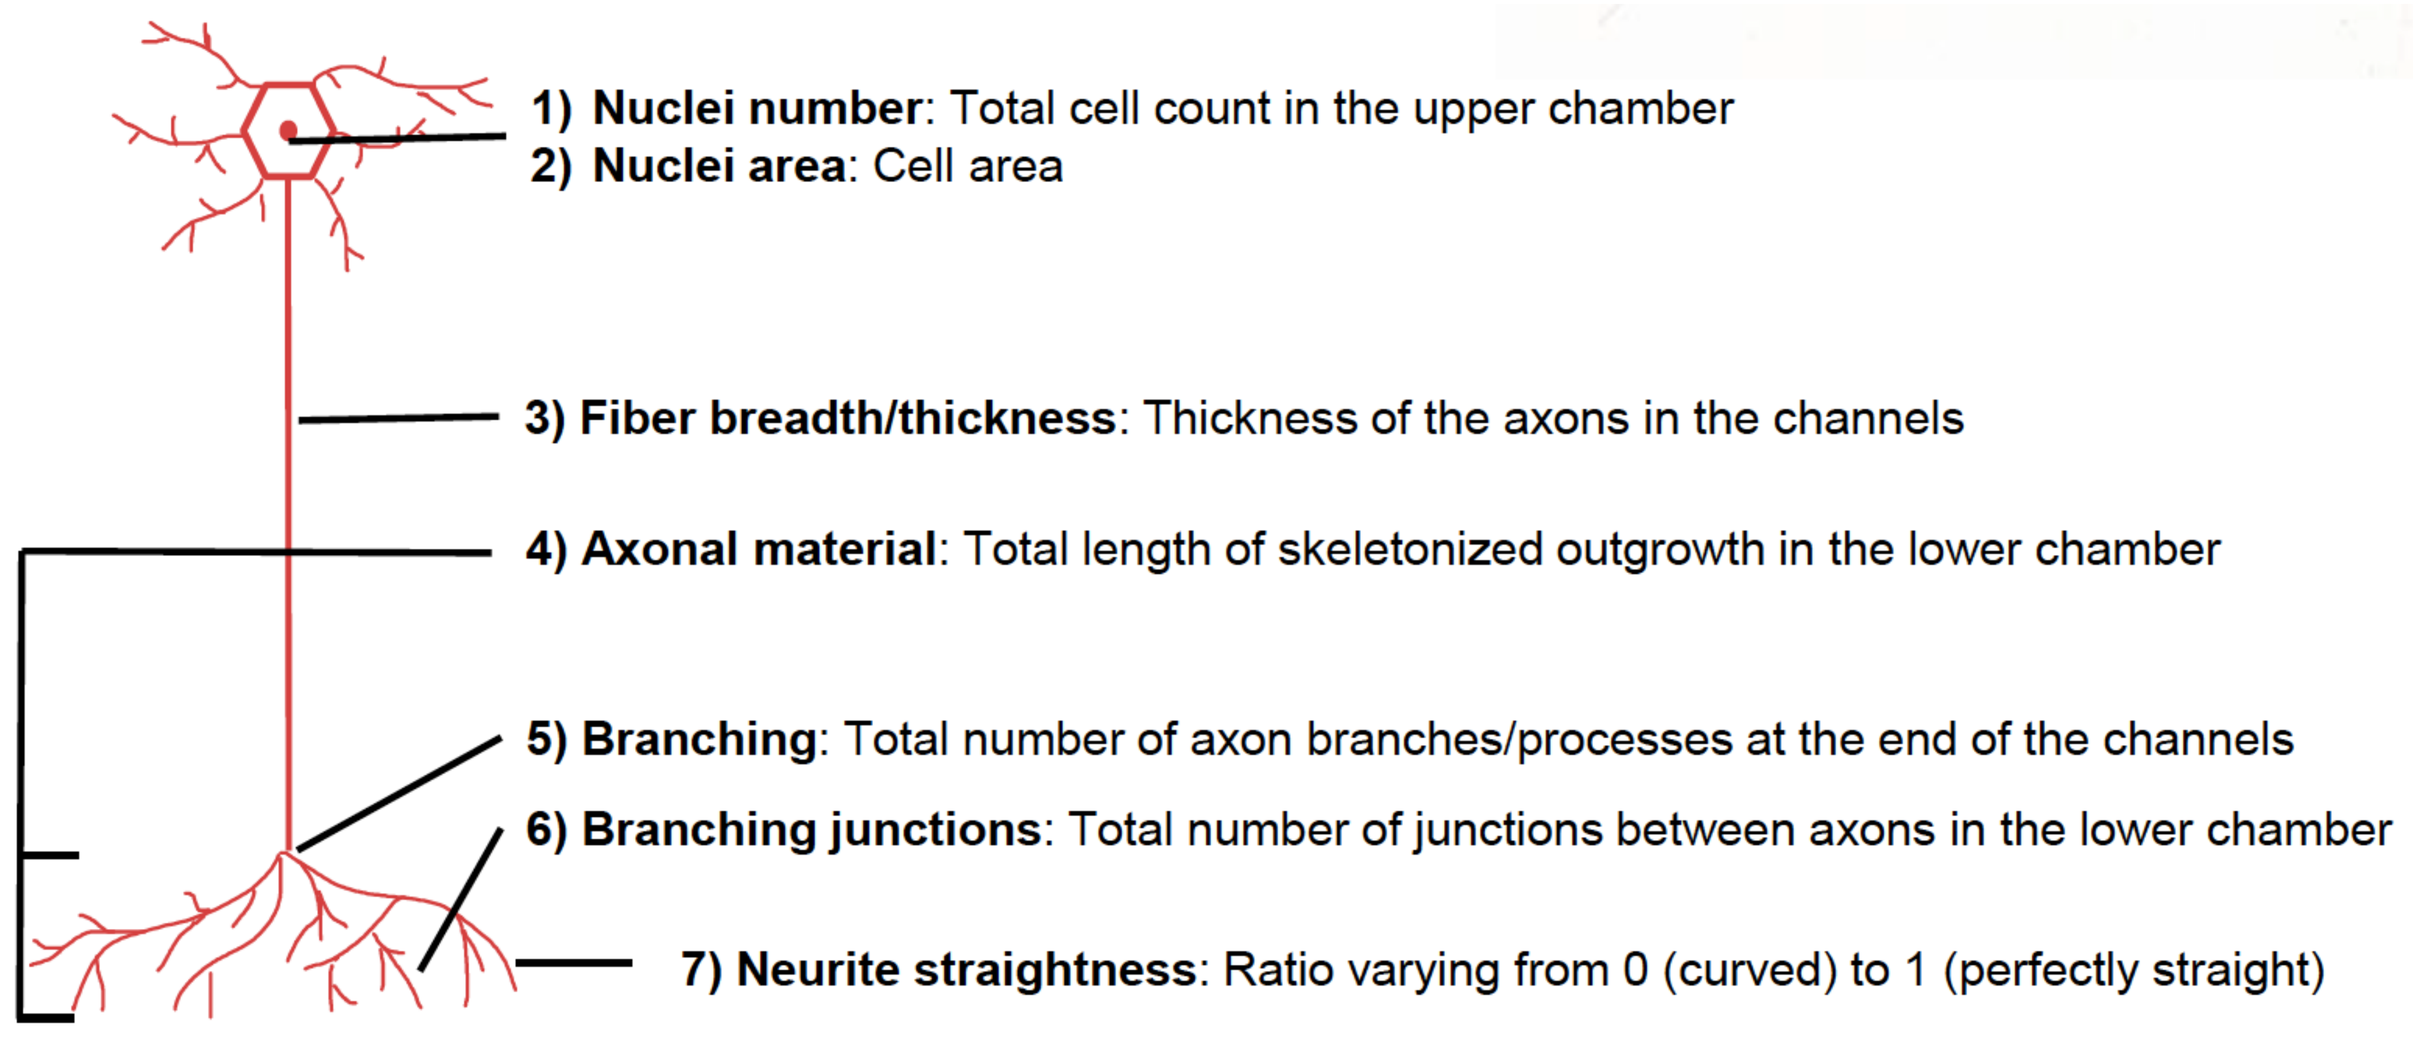
1) Nuclei number: Total cell count in the upper chamber
2) Nuclei area: Cell area
3) Fiber breadth/thickness: Thickness of the axons in the channels
4) Axonal material: Total length of skeletonized outgrowth in the lower chamber
5) Branching: Total number of axon branches/processes at the end of the channels
6) Branching junctions: Total number of junctions between axons in the lower chamber
7) Neurite straightness: Ratio varying from 0 (curved) to 1 (perfectly straight)

Figure 2. *Parameters of neuromorphological profiling performed to assess the neuroregenerative effects of HER-096 inn axotomized motor neurons.*

CONCLUSIONS

A multi-omics approach was employed to study the multimodal mechanism underlying the neuroprotective and neuroregenerative effects of HER-096 in chronic and acute neuronal degeneration. Our results suggest that HER-096 promotes functional recovery and regeneration of stressed neurons via multiple complementary ways which extend beyond modulation of UPR signaling.

RESULTS

HER-096 Shows Neuroprotective Activity *In Vivo* That is Accompaned by Normalization of Striatal Dopamine Levels and Improvement of Motor Symptoms

HER-096 administered s.c. 3 times per week in the aged mouse model of synucleinopathy protected dopamine neurons (Figure 3A) and reduced α-synuclein aggregation (Figure 3B) and phosphorylation (Figure 3E). This was accompanied by normalized striatal levels of dopamine (Figure 3C) correlating with improved motor function [bar walking test, grid walking test (Figure 3G)]. Protein levels of tyrosine hydroxylase (not shown) and dopamine transporter (Figure 3F) were increased supporting the increased number of dopamine neurons.

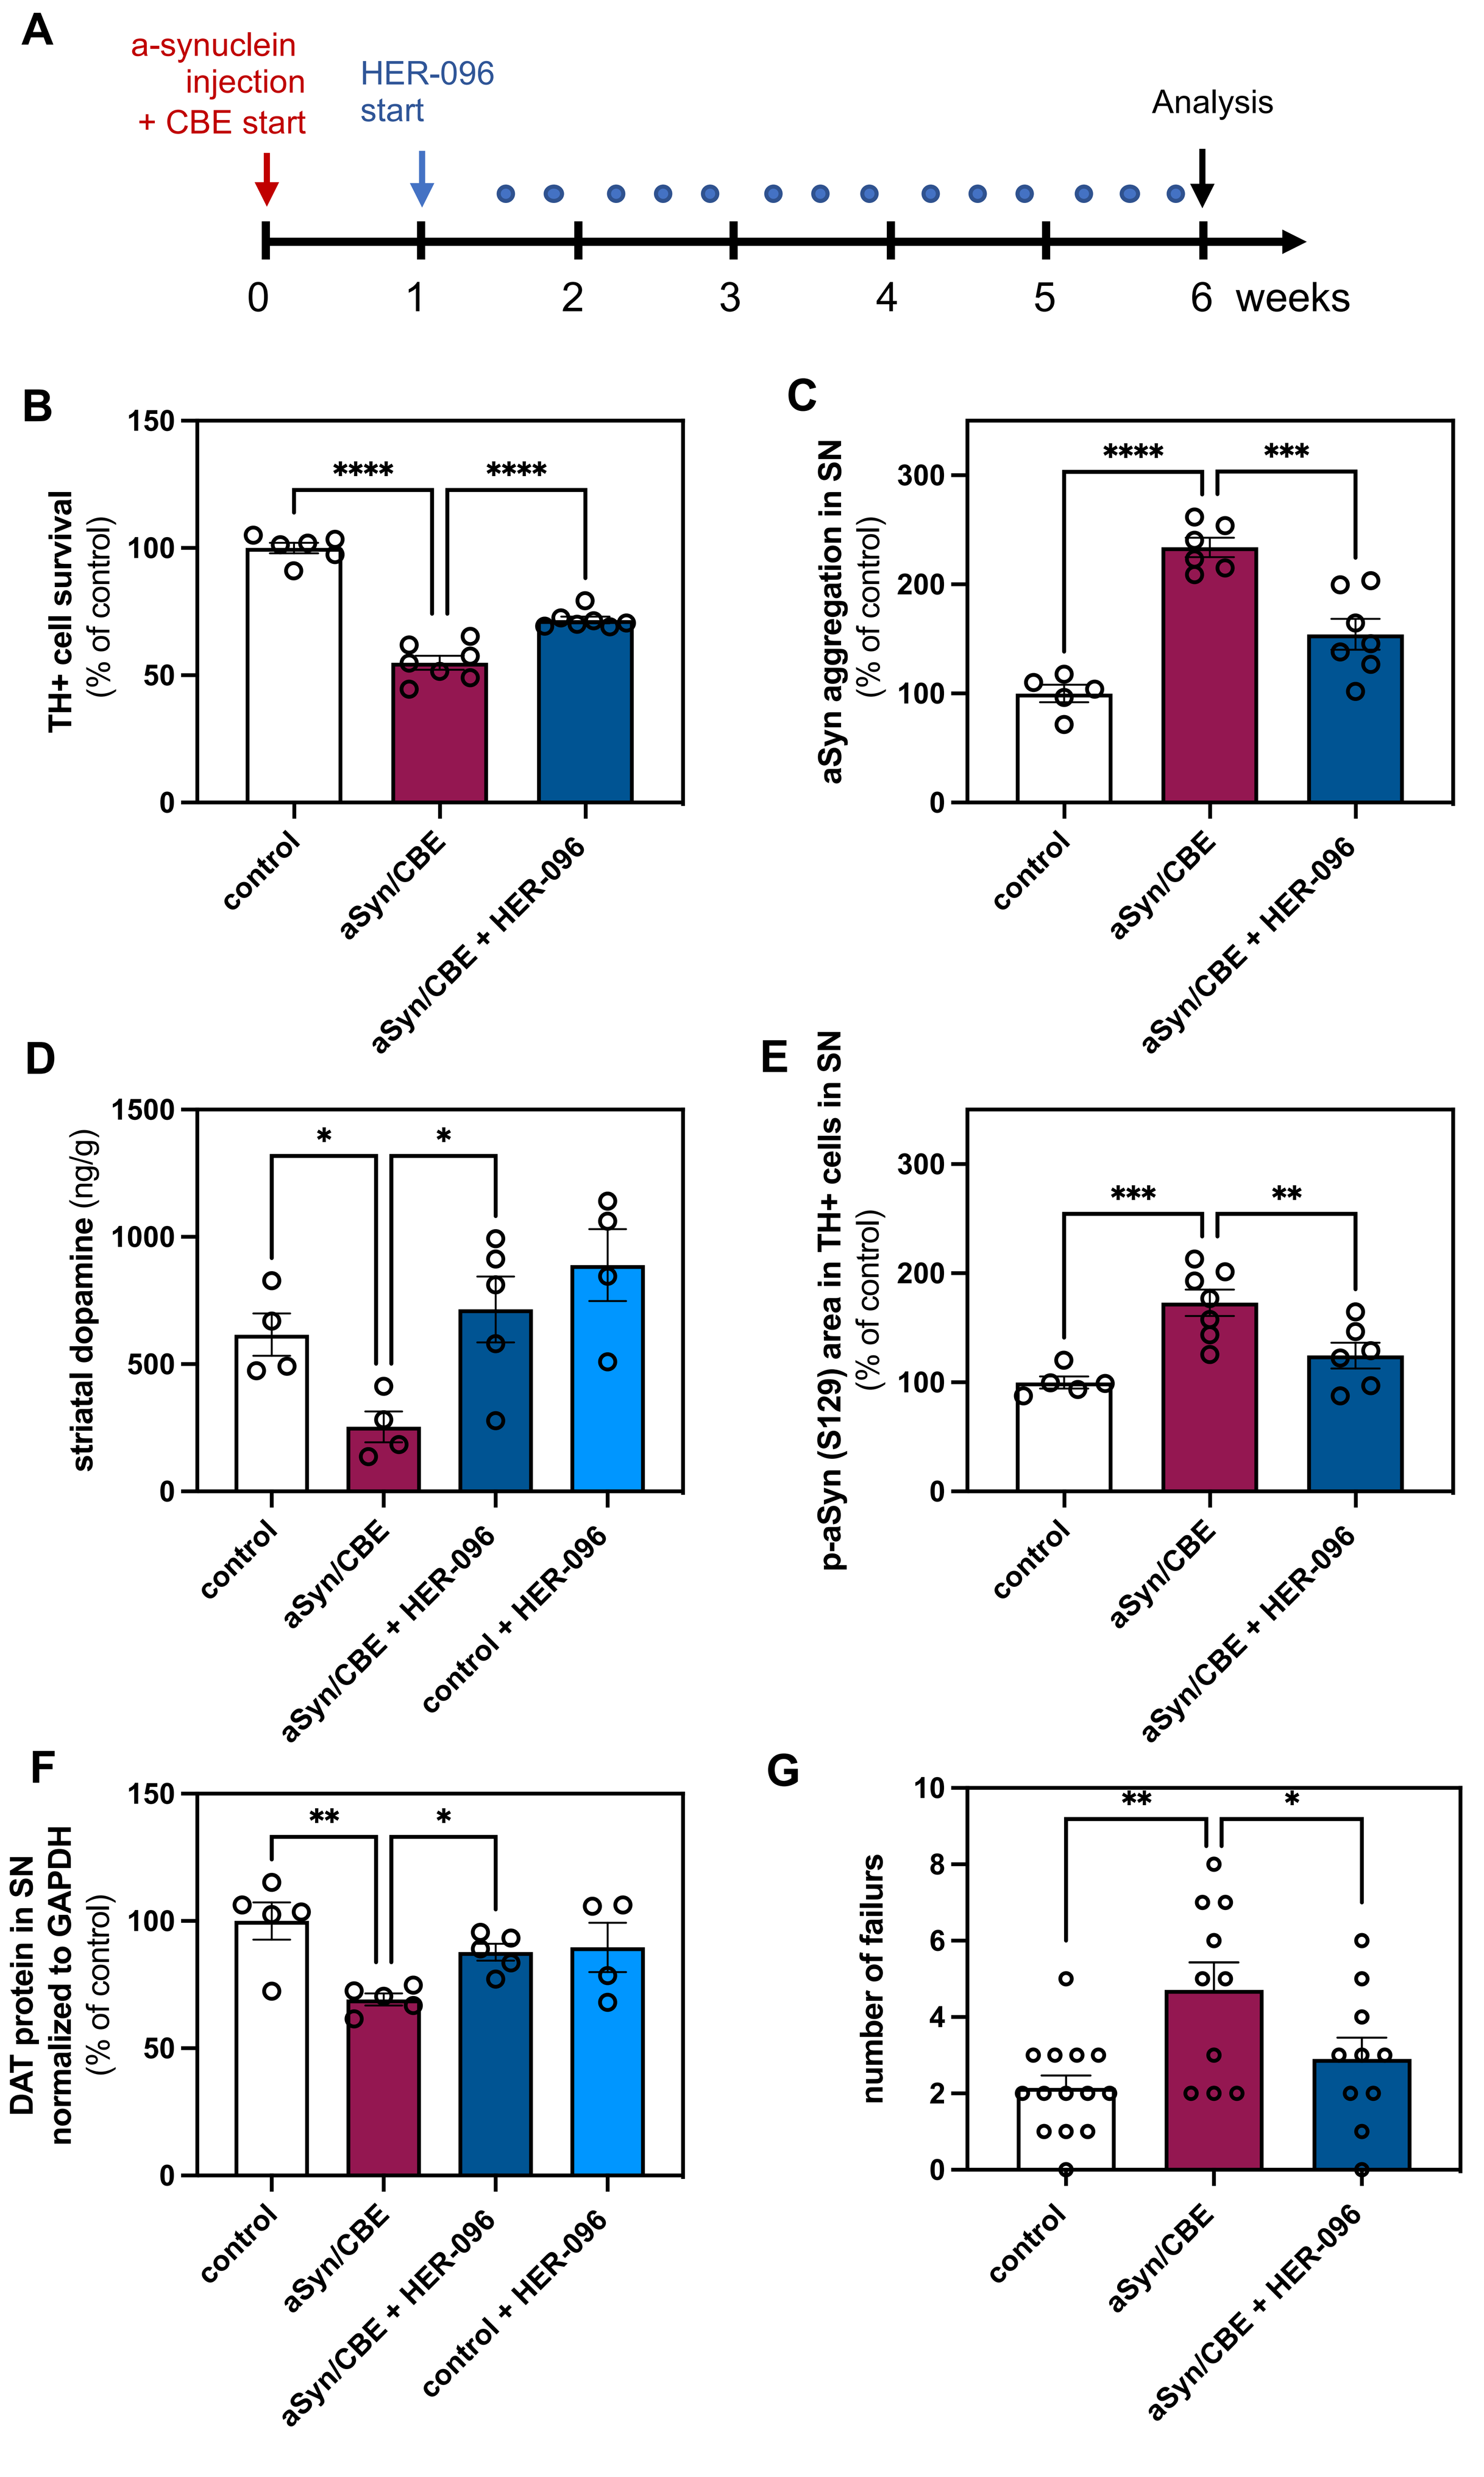
A
a-synuclein injection + CBE start
HER-096 start
Analysis
0 1 2 3 4 5 6 weeks
B
TH+ cell survival (% of control)
C
aSyn aggregation in SN (% of control)
D
striatal dopamine (ng/g)
E
p-aSyn (S129) area in TH+ cells in SN (% of control)
F
DAT protein in SN normalized to GAPDH (% of control)
G
number of failurs
control
aSyn/CBE
aSyn/CBE + HER-096
control + HER-096

Figure 3. *HER-096 was administrated to mice at 10 mg/kg s.c. three times per week for five weeks starting one week after model induction. The effects of HER-096 on dopaminergic cell survival (**B**) and on aggregated (**C**) and phosphorylated (**E**) α-synuclein were determined by IHC. (**D**) Dopamine level in striatal homogenates was assessed by LC-MS. (**F**) Effect of HER-096 treatment on protein level of dopamine transporter (DAT) in SNc. (**G**) Number of foot misplacement (failures) in the grid walking test after 2 weeks of treatment with HER-096. N=5-12. *p<0.05, **p<0.01, ***p<0.001, one-way ANOVA followed by uncorrected Fisher's LSD test for pairwise comparison of mean dose effect versus model group treated with vehicle (aSyn/CBE). Results are presented as a mean percentage from the control level +/- SEM.*

HER-096 Treatment Has Broad Effects on Cellular Proteostasis and Viability Based on Gene Expression Analysis

Gene expression changes in the midbrain bulk striatum and substantia nigra tissue of aged synycleinopathy mice treated with s.c. HER-096 were assessed by Affymetrix microarray and geneset enrichment analysis (GSEA). The GSEA comparions between healthy aged mice and the synucleinopathy model strongly support a Parkinson's disease-like degenerative process taking place in the nigrostriatal system (Figure 4A), highlighting the ER and mitochondria cellular compartments mostly affected.

Comparison of HER-096 vs vehicle-treated synucleinopathy mice suggest that HER-096 has induced broad changes affecting several critical cellular functions involved in neurodegeneration, such as proteostasis control via the proteasome and ribosomes, as well as mitochondrial function (Figure 4B). This data suggests that modulation of UPR by HER-096 in degenerating nigrostriatal pathway affects multiple cellular systems beyond the ER that likely contribute to the functional recovery shown in Figure 3.

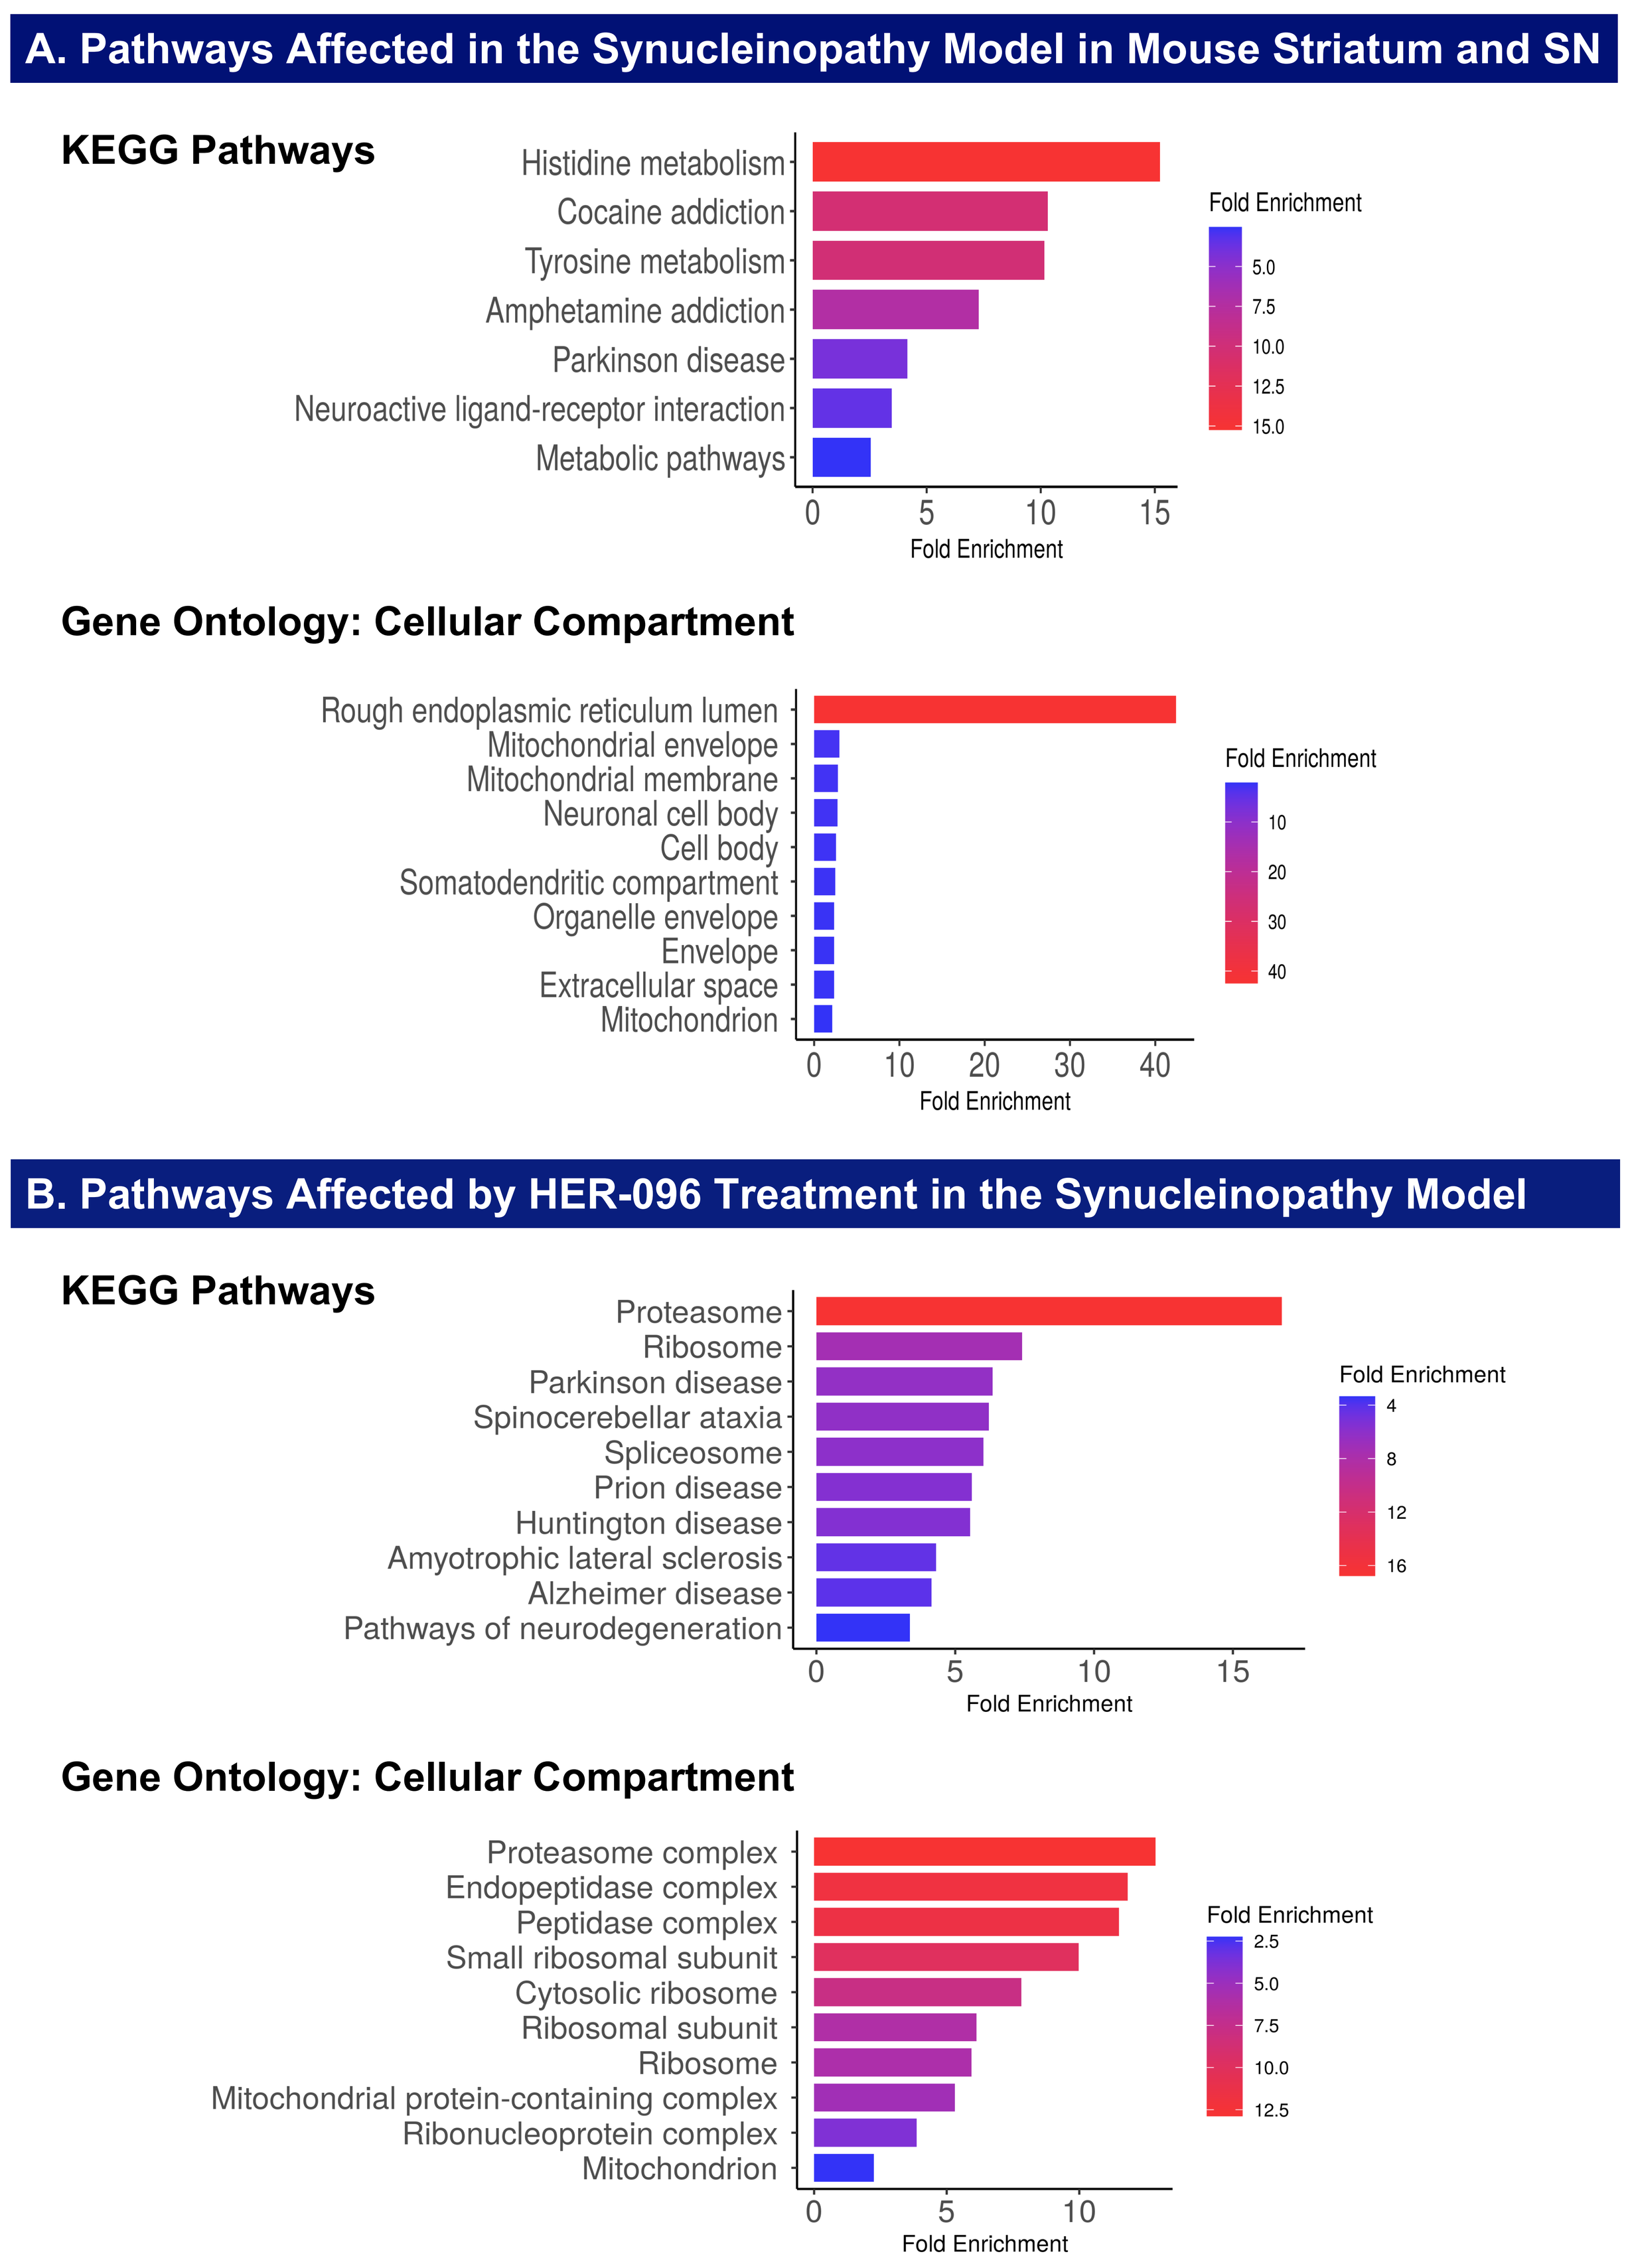
A. Pathways Affected in the Synucleinopathy Model in Mouse Striatum and SN
KEGG Pathways
Histidine metabolism
Cocaine addiction
Tyrosine metabolism
Amphetamine addiction
Parkinson disease
Neuroactive ligand-receptor interaction
Metabolic pathways
Fold Enrichment
Gene Ontology: Cellular Compartment
Rough endoplasmic reticulum lumen
Mitochondrial envelope
Mitochondrial membrane
Neuronal cell body
Cell body
Somatodendritic compartment
Organelle envelope
Envelope
Extracellular space
Mitochondrion
Fold Enrichment
B. Pathways Affected by HER-096 Treatment in the Synucleinopathy Model
KEGG Pathways
Proteasome
Ribosome
Parkinson disease
Spinocerebellar ataxia
Spliceosome
Prion disease
Huntington disease
Amyotrophic lateral sclerosis
Alzheimer disease
Pathways of neurodegeneration
Fold Enrichment
Gene Ontology: Cellular Compartment
Proteasome complex
Endopeptidase complex
Peptidase complex
Small ribosomal subunit
Cytosolic ribosome
Ribosomal subunit
Ribosome
Mitochondrial protein-containing complex
Ribonucleoprotein complex
Mitochondrion
Fold Enrichment

Figure 4. *Gene set enrichment analysis (GSEA) for differently expressed genes in the dissected striatum+substantia nigra. Analysis was done separately for pathways affected by the synucleinopathy model (vs healthy aged animals) (**A**) and after a 5-week HER-096 treatment in the synucleinopathy model animals (vs vehicle treatment)(**B**).*

HER-096 Demonstrates Neuroregenerative Effects in Axotomized hiPSC Motor Neurons

Incubation with HER-096 for 6 days resulted in increased total length of skeletonized outgrowth (axonal material), total number of axonal branches and branch junctions in the axotomized hiPSC motor neurons (Figure 5). HER-096 did not significantly affect neuromorphological parameters in non-axotomized motor neurons.

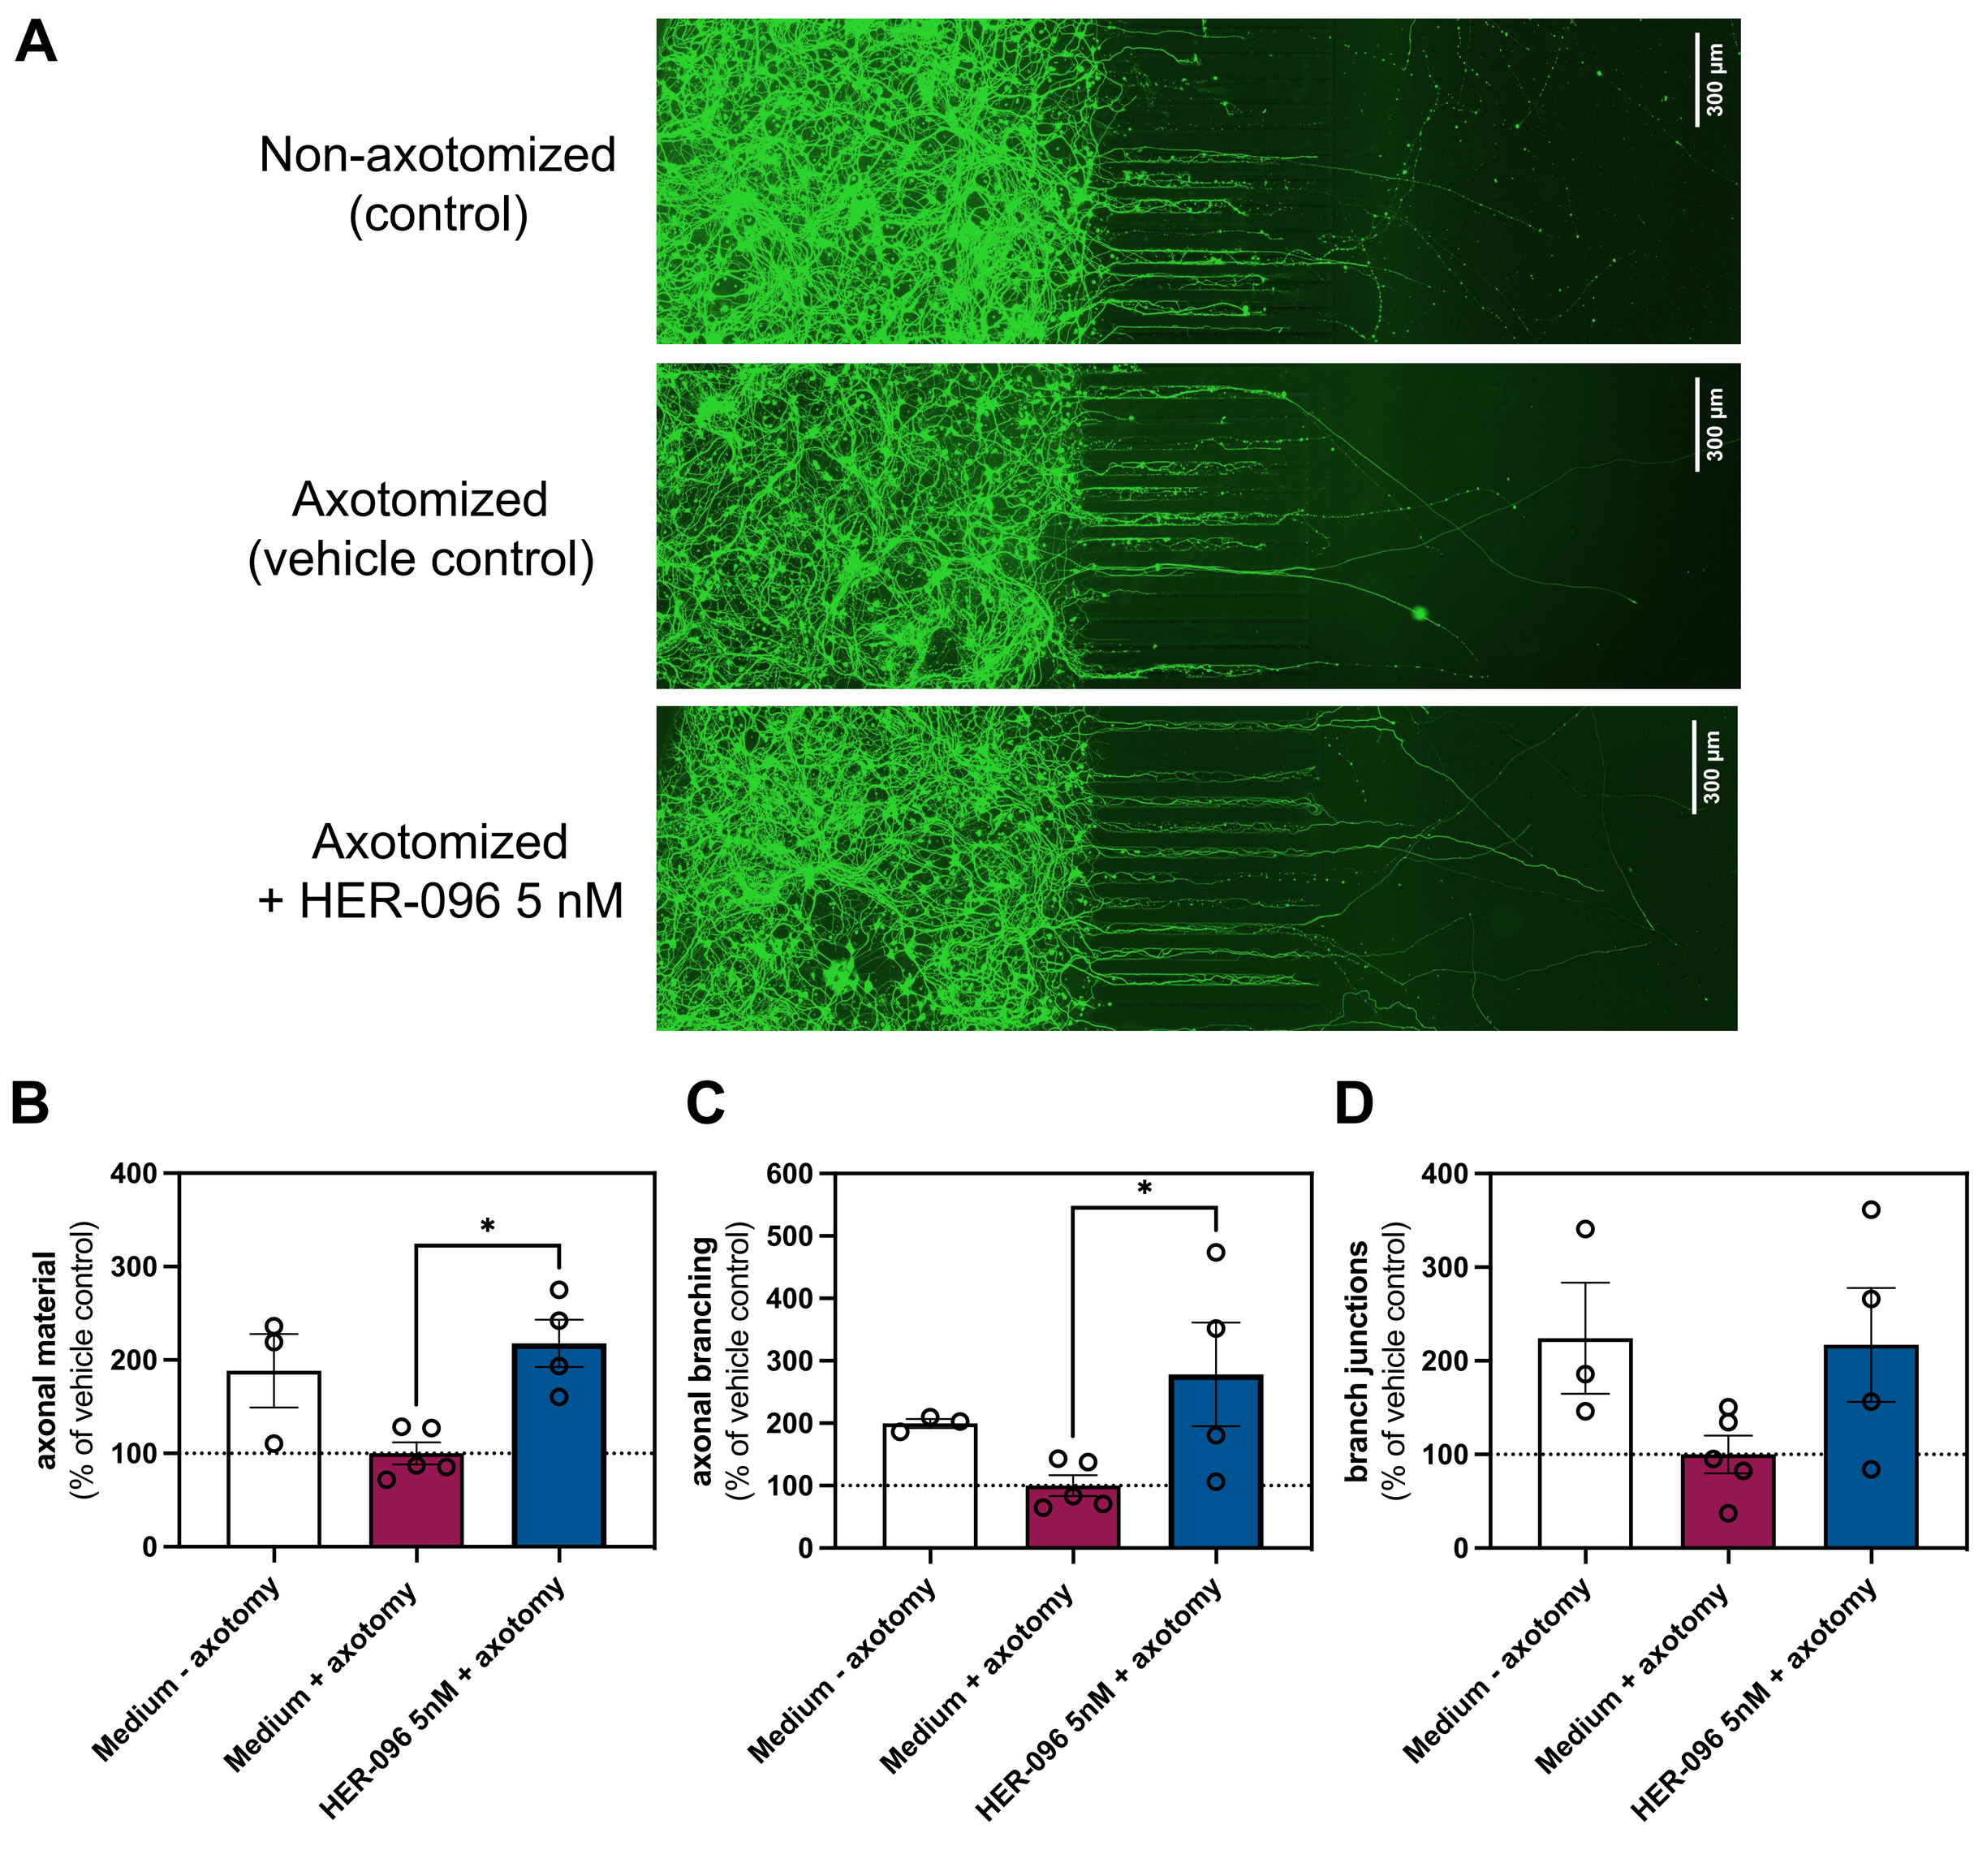
A
Non-axotomized (control)
Axotomized (vehicle control)
Axotomized + HER-096 5 nM
300 µm
B
axonal material (% of vehicle control)
C
axonal branching (% of vehicle control)
D
branch junctions (% of vehicle control)
Medium - axotomy
Medium + axotomy
HER-096 5nM + axotomy

Figure 5. (A) *Representative images of motor neurons in microfluidic chambers in healthy/control condition, after axotomy, and after axotomy and 6-days treatment with HER-096 5 nM. Effect of HER-096 treatment on neuromorphological parameters of axotomized motor neurons: total length of skeletomized outgrowth (axonal material)* ***(B)****, total number of axonal branches* ***(C)*** *and branch junctions* ***(D)*** *normalized to the to total number of cells. N=3-5. *p<0.05, Kruskal-Wallis with uncorrected Dunn's test for pairwise comparison versus vehicle-treated group (medium + axotomy). Results are presented as a mean percentage from the vehicle level +/- SEM.*

European Innovation Council
Co-funded by the European Union

HERANTIS PHARMA

www.herantis.com